Improved Cognitive Processing Speed With Ublituximab in Patients With Highly Active Relapsing Multiple Sclerosis

Derrick Robertson, MD,[1] Lawrence Steinman, MD,[2] Edward J. Fox, MD, PhD,[3] Hans-Peter Hartung, MD,[4-7] Enrique Alvarez, MD, PhD,[8] Peiqing Qian, MD,[9] Sibyl Wray, MD,[10] DeRen Huang, MD, PhD,[11] Krzysztof Selmaj, MD, PhD,[12,13] Daniel Wynn, MD,[14] Koby Mok, PhD,[15] Christopher A. Garner, PA-C,[15] Bruce A. C. Cree, MD, PhD, MAS[16]

[1]University of South Florida, Tampa, FL; [2]Stanford University, Stanford, CA; [3]Central Texas Neurology Consultants, Round Rock, TX; [4]Heinrich Heine University Düsseldorf, Düsseldorf, Germany; [5]Brain and Mind Centre, University of Sydney, Sydney, Australia; [6]Medical University of Vienna, Vienna, Austria; [7]Palacký University Olomouc, Olomouc, Czech Republic; [8]University of Colorado, Aurora, CO; [9]Swedish Medical Center, Seattle, WA; [10]Hope Neurology, Knoxville, TN; [11]Center for Multiple Sclerosis, Mount Carmel Health System, Westerville, OH; [12]Center of Neurology, Lodz, Poland; [13]University of Warmia and Mazury, Olsztyn, Poland; [14]Consultants in Neurology, Northbrook, IL; [15]TG Therapeutics, New York, NY; [16]UCSF Weill Institute for Neurosciences, University of California San Francisco, San Francisco, CA

OBJECTIVE

- To evaluate changes in cognitive processing speed using the Symbol Digit Modalities Test (SDMT) in participants with relapsing multiple sclerosis (RMS) in the ULTIMATE I and ULTIMATE II studies

KEY FINDINGS

- In a pooled post hoc analysis, ublituximab was associated with a mean 4.1 increase in SDMT score at Week 96 from baseline, established as a clinically meaningful improvement[1]
- In previously treated participants with highly active disease at baseline, the mean change from baseline was 2.8 vs 1.2, 3.8 vs 2.7, and 5.0 vs 2.5 for ublituximab vs teriflunomide at Weeks 24, 48, and 96, respectively. The difference at Week 96 was statistically significant: *P*=0.0205

CONCLUSIONS

- Participants with RMS receiving ublituximab in ULTIMATE I and II had a clinically meaningful improvement in cognitive processing speed as measured by the SDMT at 96 weeks
- In previously treated participants with highly active disease at baseline, ublituximab treatment provided clinically meaningful and statistically significant improvement in cognitive processing speed (SDMT score) vs teriflunomide at 96 weeks

REFERENCES 1. Benedict RHB, et al. *Mult Scler*. 2017;23(5)721-733. **2.** Le Garff-Tavernier M, et al. *Leukemia*. 2014;28(1):230-233. **3.** Babiker HM, et al. *Expert Opin Investig Drugs*. 2018;27(4):407-412. **4.** Steinman L, et al. Presented at: ECTRIMS; October 13-15, 2021; Virtual. Oral presentation 117. **5.** Ferrara C, et al. *Proc Natl Acad Sci U S A*. 2011;108(31):12669-12674. **6.** Sun Y, et al. *J Biol Chem*. 2021;297(1):100826. **7.** de Romeuf C, et al. *Br J Haematol*. 2008;140(6):635-643. **8.** Fox E, et al. *Mult Scler*. 2021;27(3):420-429. **9.** Arrambide G, et al. *Mult Scler*. 2020;26(9):1045-1063.

ACKNOWLEDGMENTS The authors thank the participants and their families for participating in the ULTIMATE I and II studies. The authors also thank Apollo Medical Communications for providing medical writing and editorial support, which was funded by TG Therapeutics. The ULTIMATE I and II studies were sponsored by TG Therapeutics.

Copies of this poster obtained by QR code are for reference only and may not be reused as a whole or in part without the permission of the authors and study sponsor.

BACKGROUND

- Ublituximab is a novel monoclonal antibody that targets a unique epitope of CD20 and is glycoengineered for enhanced antibody-dependent cellular cytotoxicity **(Figure 1)**[2,3]
- Ublituximab is administered in lower doses and with shorter infusion times compared with other currently infused anti-CD20 therapies[4]
- ULTIMATE I (NCT03277261) and ULTIMATE II (NCT03277248) are identical, Phase 3, randomized, multicenter, double-blind, active-control, double-dummy studies evaluating the efficacy and safety of ublituximab vs teriflunomide in participants with RMS[4]
- ULTIMATE I and II met their primary endpoint, demonstrating a statistically significant reduction in annualized relapse rate for ublituximab compared with teriflunomide as well as significant improvements in the number of gadolinium-enhancing (Gd+) T1 lesions and the number of new/enlarging T2 lesions[4]
- SDMT is a reliable and validated tool for evaluating slowed cognitive processing speed commonly seen in people with RMS. A 4-point increase has been established as a clinically meaningful improvement in cognitive processing[1]

Figure 1. Ublituximab Is Glycoengineered to Enhance ADCC

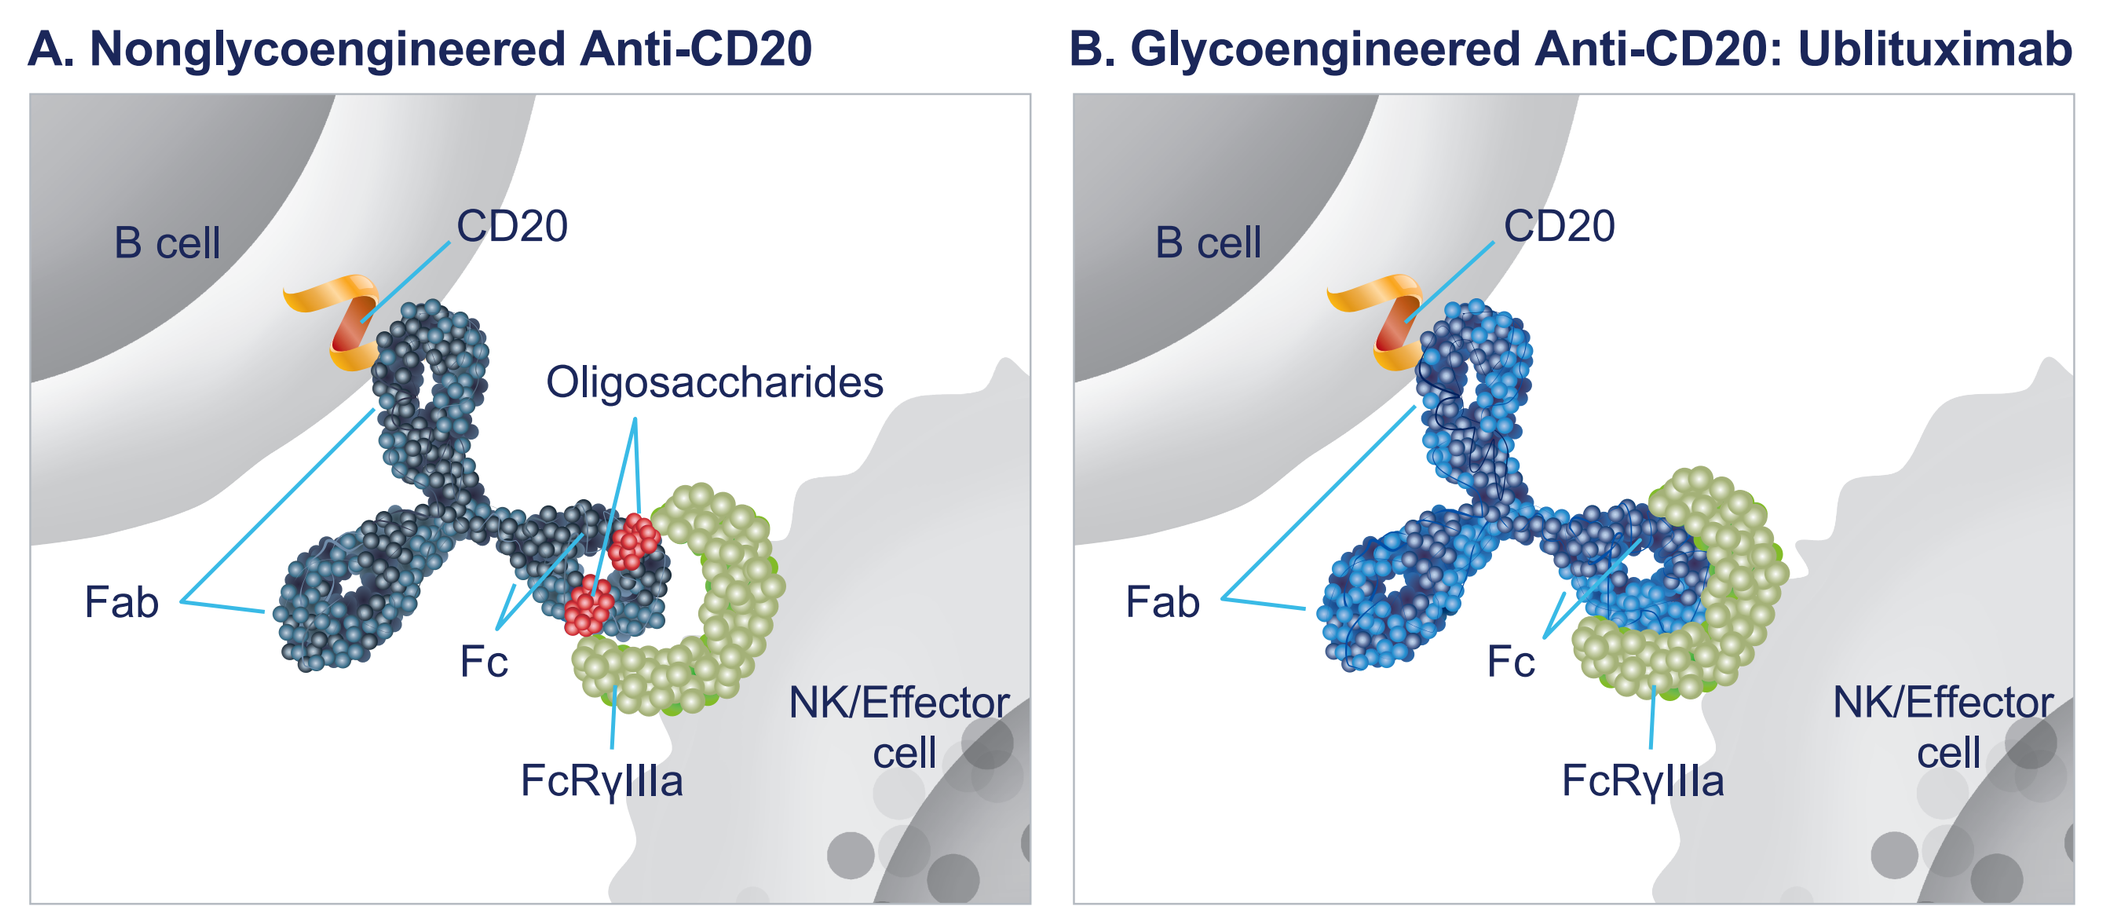
A. Nonglycoengineered Anti-CD20
B cell
CD20
Oligosaccharides
Fab
Fc
FcRγIIIa
NK/Effector cell
B. Glycoengineered Anti-CD20: Ublituximab
B cell
CD20
Fab
Fc
FcRγIIIa
NK/Effector cell

(A) In nonglycoengineered anti-CD20 antibodies, the core fucose of Fc-linked oligosaccharides sterically blocks interaction with FcγRIIIa, reducing affinity.[5,6]
(B) Ublituximab is glycoengineered to have a low fucose content in the Fc region, which allows for closer interaction and enhanced affinity for all variants of FcγRIIIa.[6-8]

ADCC, antibody-dependent cellular cytotoxicity; NK, natural killer.

METHODS

- ULTIMATE I and II enrolled a total of 1094 adult participants from 10 countries with a diagnosis of RMS (relapsing-remitting or secondary-progressive) with disease activity[4]
- Participants received ublituximab 450 mg administered by 1-hour intravenous infusion every 24 weeks (following Day 1 infusion of 150 mg and Day 15 infusion of 450 mg) or teriflunomide 14 mg oral once daily for 96 weeks[4]
- SDMT evaluations were performed at baseline and Weeks 24, 48, and 96
- Pooled SDMT data from both studies were evaluated in post hoc analyses of the modified intention-to-treat (mITT) population and the subpopulation of participants with highly active disease, defined as ≥1 relapse in prior year and either ≥1 Gd+ T1 lesion or ≥9 T2 lesions at baseline[9]
- Participants with highly active disease included in the analysis received an approved disease-modifying therapy prior to study enrollment

RESULTS

- Baseline characteristics for the overall mITT population and the subpopulation of participants with high disease activity and prior treatment are shown in **Table 1**
- In the mITT population, ublituximab was associated with a mean 4.1 increase in SDMT score from baseline at Week 96, established as a clinically meaningful improvement.[1] The differences in SDMT score were not significant vs teriflunomide **(Figures 2 and 3)**
- In participants with highly active disease at baseline, the mean baseline SDMT score was 49.8 and 48.2 for the ublituximab (n=181) and teriflunomide (n=158) groups, respectively
- The mean change from baseline in SDMT score was 2.8 vs 1.2, 3.8 vs 2.7, and 5.0 vs 2.5 for ublituximab vs teriflunomide at Weeks 24, 48, and 96, respectively **(Figure 4)**
- The difference in mean change from baseline at Week 96 for ublituximab vs teriflunomide was statistically significant: *P*=0.0205 **(Figure 5)**

Table 1. Participant Demographics and Baseline Characteristics

Characteristic *Mean ± standard deviation or %*	Overall Population		Highly Active Disease With Prior Treatment[a]	
	Teriflunomide (n=546)	Ublituximab (n=543)	Teriflunomide (n=158)	Ublituximab (n=181)
Age, years	36.6±9.3	35.3±8.6	38.4±9.4	35.5±8.5
Sex, female, %	65.0	63.4	69.6	66.3
Duration of MS since first symptoms, years	7.1±6.1	7.4±6.5	9.3±6.4	9.2±6.4
Number of relapses in last 12 months	1.3±0.7	1.3±0.7	1.4±0.7	1.4±0.7
Number of relapses in last 24 months	1.9±1.0	1.8±1.0	2.2±1.2	2.0±1.2
Time since most recent relapse, months	6.2±4.8	7.1±8.5	5.9±3.2	5.8±3.1
EDSS score at screening	2.9±1.2	2.9±1.3	3.3±1.2	3.1±1.3
T2 lesion volume, cm^3	15.3±16.7	15.3±14.8	17.6±19.2	16.3±15.2
Number of T2 lesions	62.2±39.2	64.7±39.9	61.8±38.7	65.0±39.9
Participants free of Gd+ T1 lesions, %	53.3	52.3	56.3	54.7

[a]Defined as ≥1 relapse in prior year and either ≥1 Gd+ T1 lesion or ≥9 T2 lesions at baseline. Participants received approved disease-modifying therapy prior to study enrollment.
EDSS, Expanded Disability Status Scale; Gd+, gadolinium-enhancing; MS, multiple sclerosis.

Figure 2. Change in SDMT Score From Baseline to Week 96 (mITT Population)

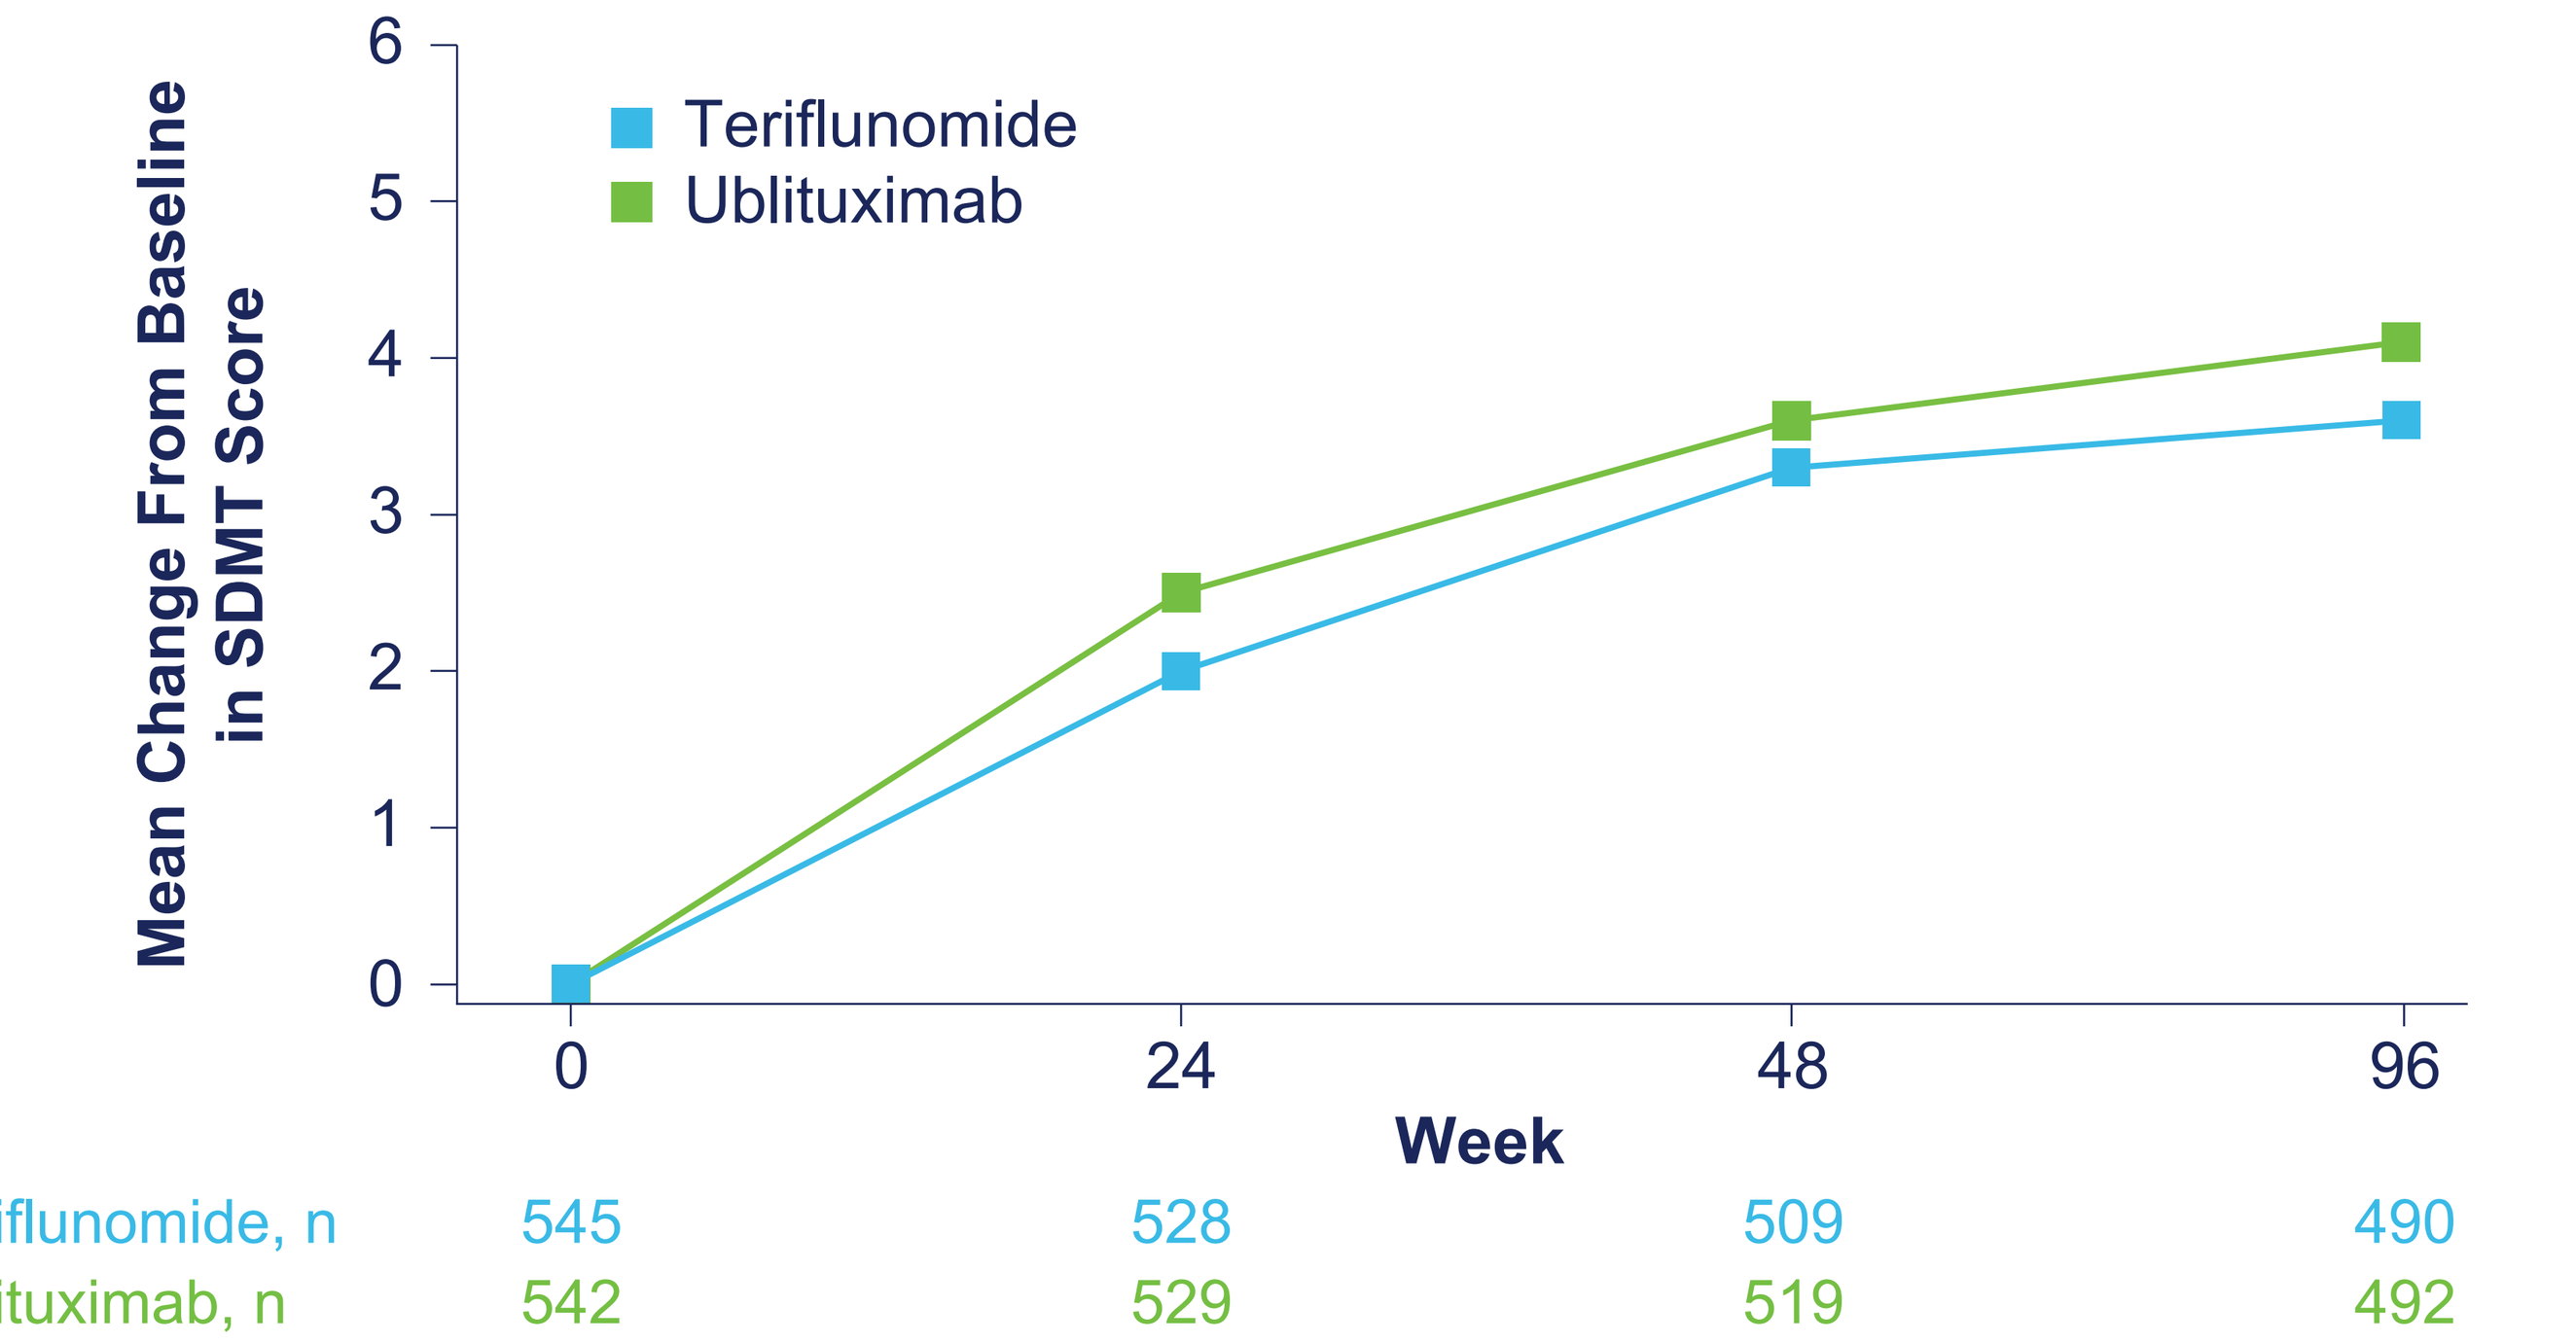
Mean Change From Baseline in SDMT Score
Teriflunomide
Ublituximab
0
1
2
3
4
5
6
0
24
48
96
Week
Teriflunomide, n 545 528 509 490
Ublituximab, n 542 529 519 492

Pooled post hoc analysis. mITT population. mITT, modified intention-to-treat; SDMT, Symbol Digit Modalities Test.

Figure 3. Change in SDMT Score From Baseline at Week 96 (mITT Population)

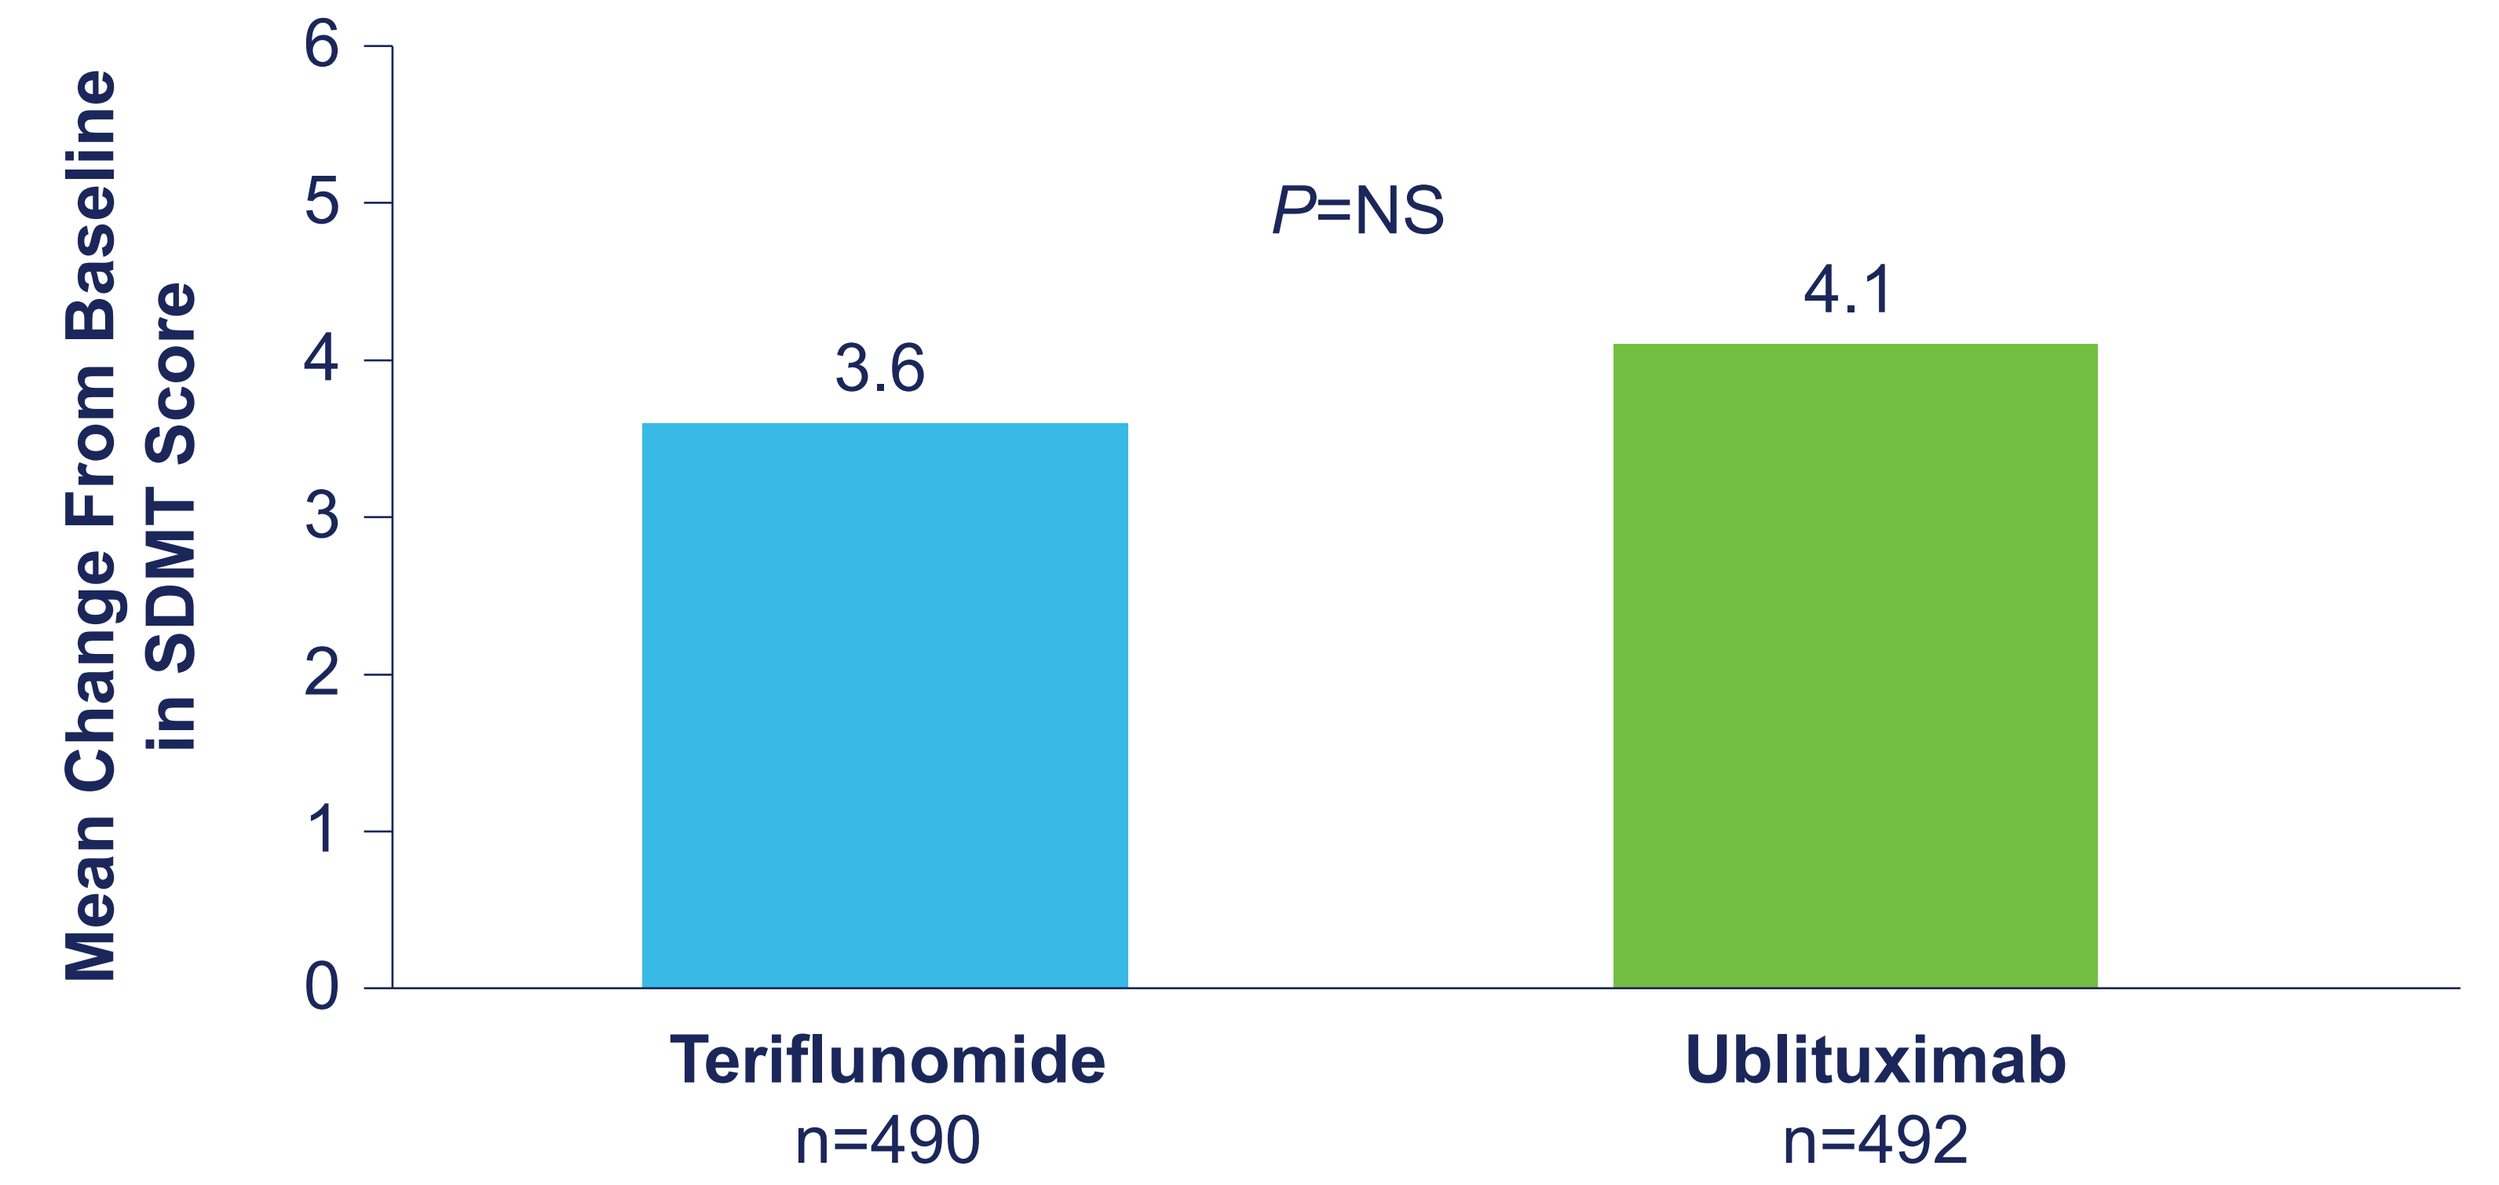
Mean Change From Baseline in SDMT Score
P=NS
3.6
4.1
Teriflunomide
n=490
Ublituximab
n=492

Pooled post hoc analysis. mITT population. mITT, modified intention-to-treat; NS, nonsignificant; SDMT, Symbol Digit Modalities Test.

Figure 4. Change in SDMT Score From Baseline to Week 96 (Participants With Highly Active Disease)[a]

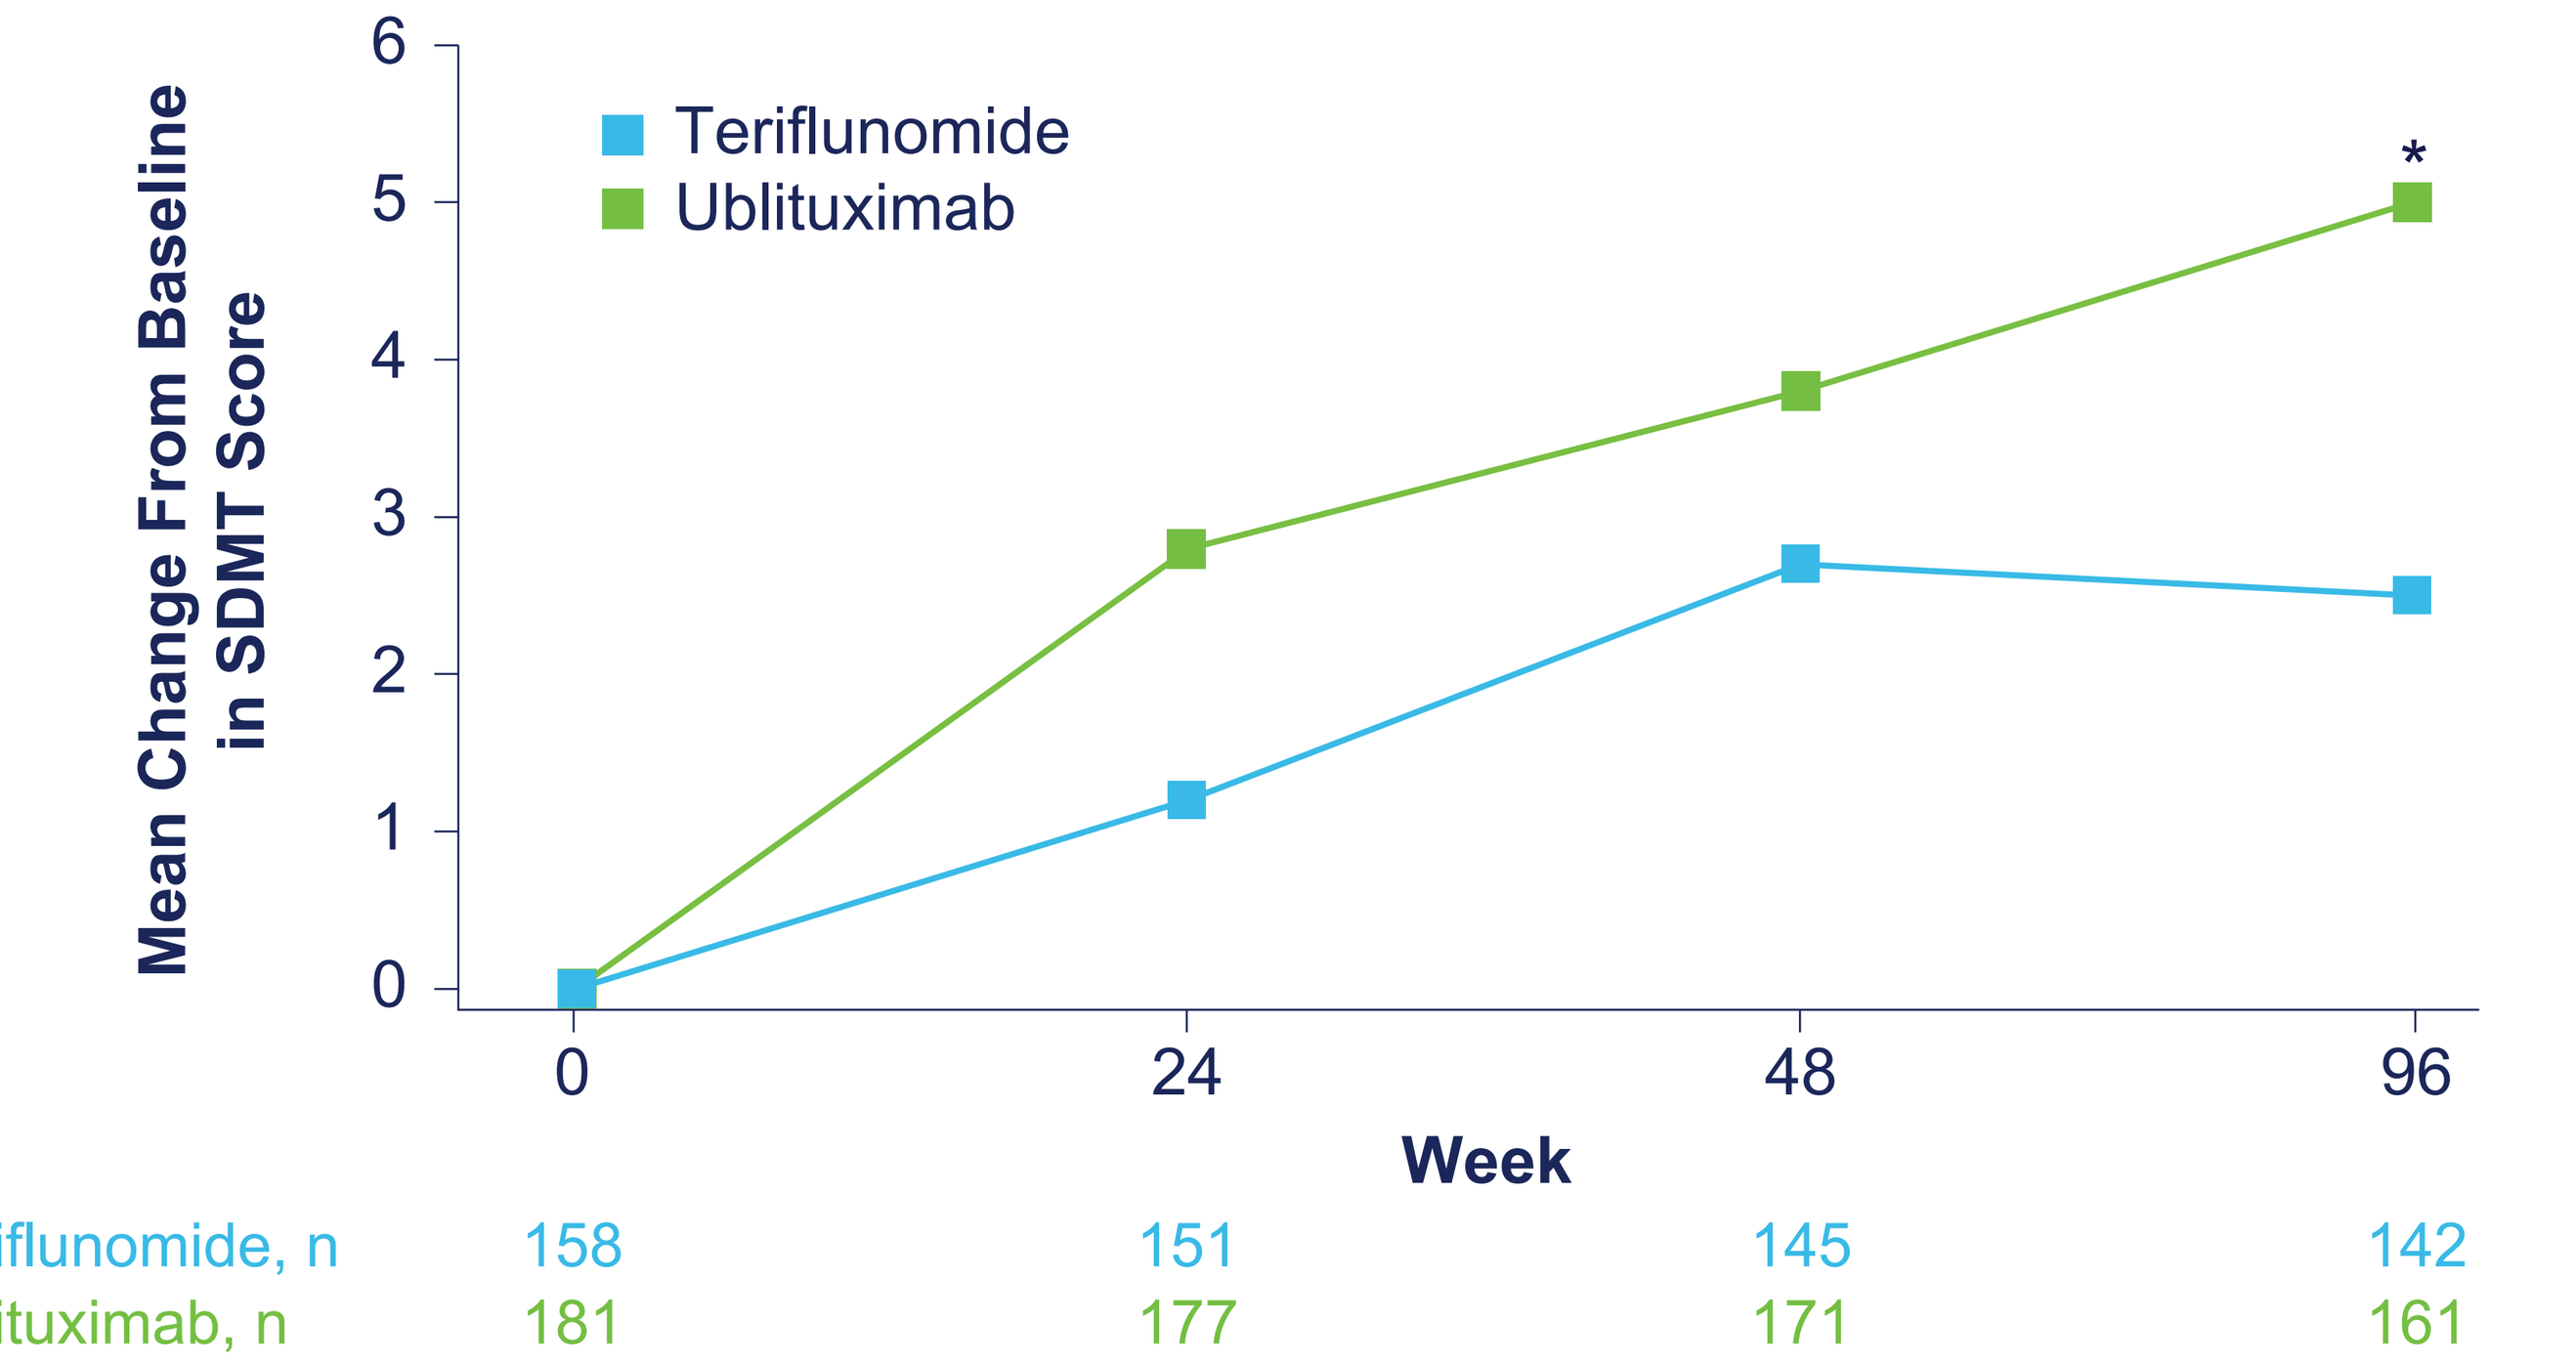
Mean Change From Baseline in SDMT Score
Teriflunomide
Ublituximab
*
0
24
48
96
Week
Teriflunomide, n 158 151 145 142
Ublituximab, n 181 177 171 161

[a]Highly active disease at baseline defined as ≥1 relapse in prior year and either ≥1 Gd+ T1 lesion or ≥9 T2 lesions at baseline. Participants received approved disease-modifying therapy prior to enrollment. **P*=0.0205.
Pooled post hoc analysis. mITT population. Gd+, gadolinium-enhancing; mITT, modified intention-to-treat; SDMT, Symbol Digit Modalities Test.

Figure 5. Change in SDMT Score From Baseline at Week 96 (Participants With Highly Active Disease)[a]

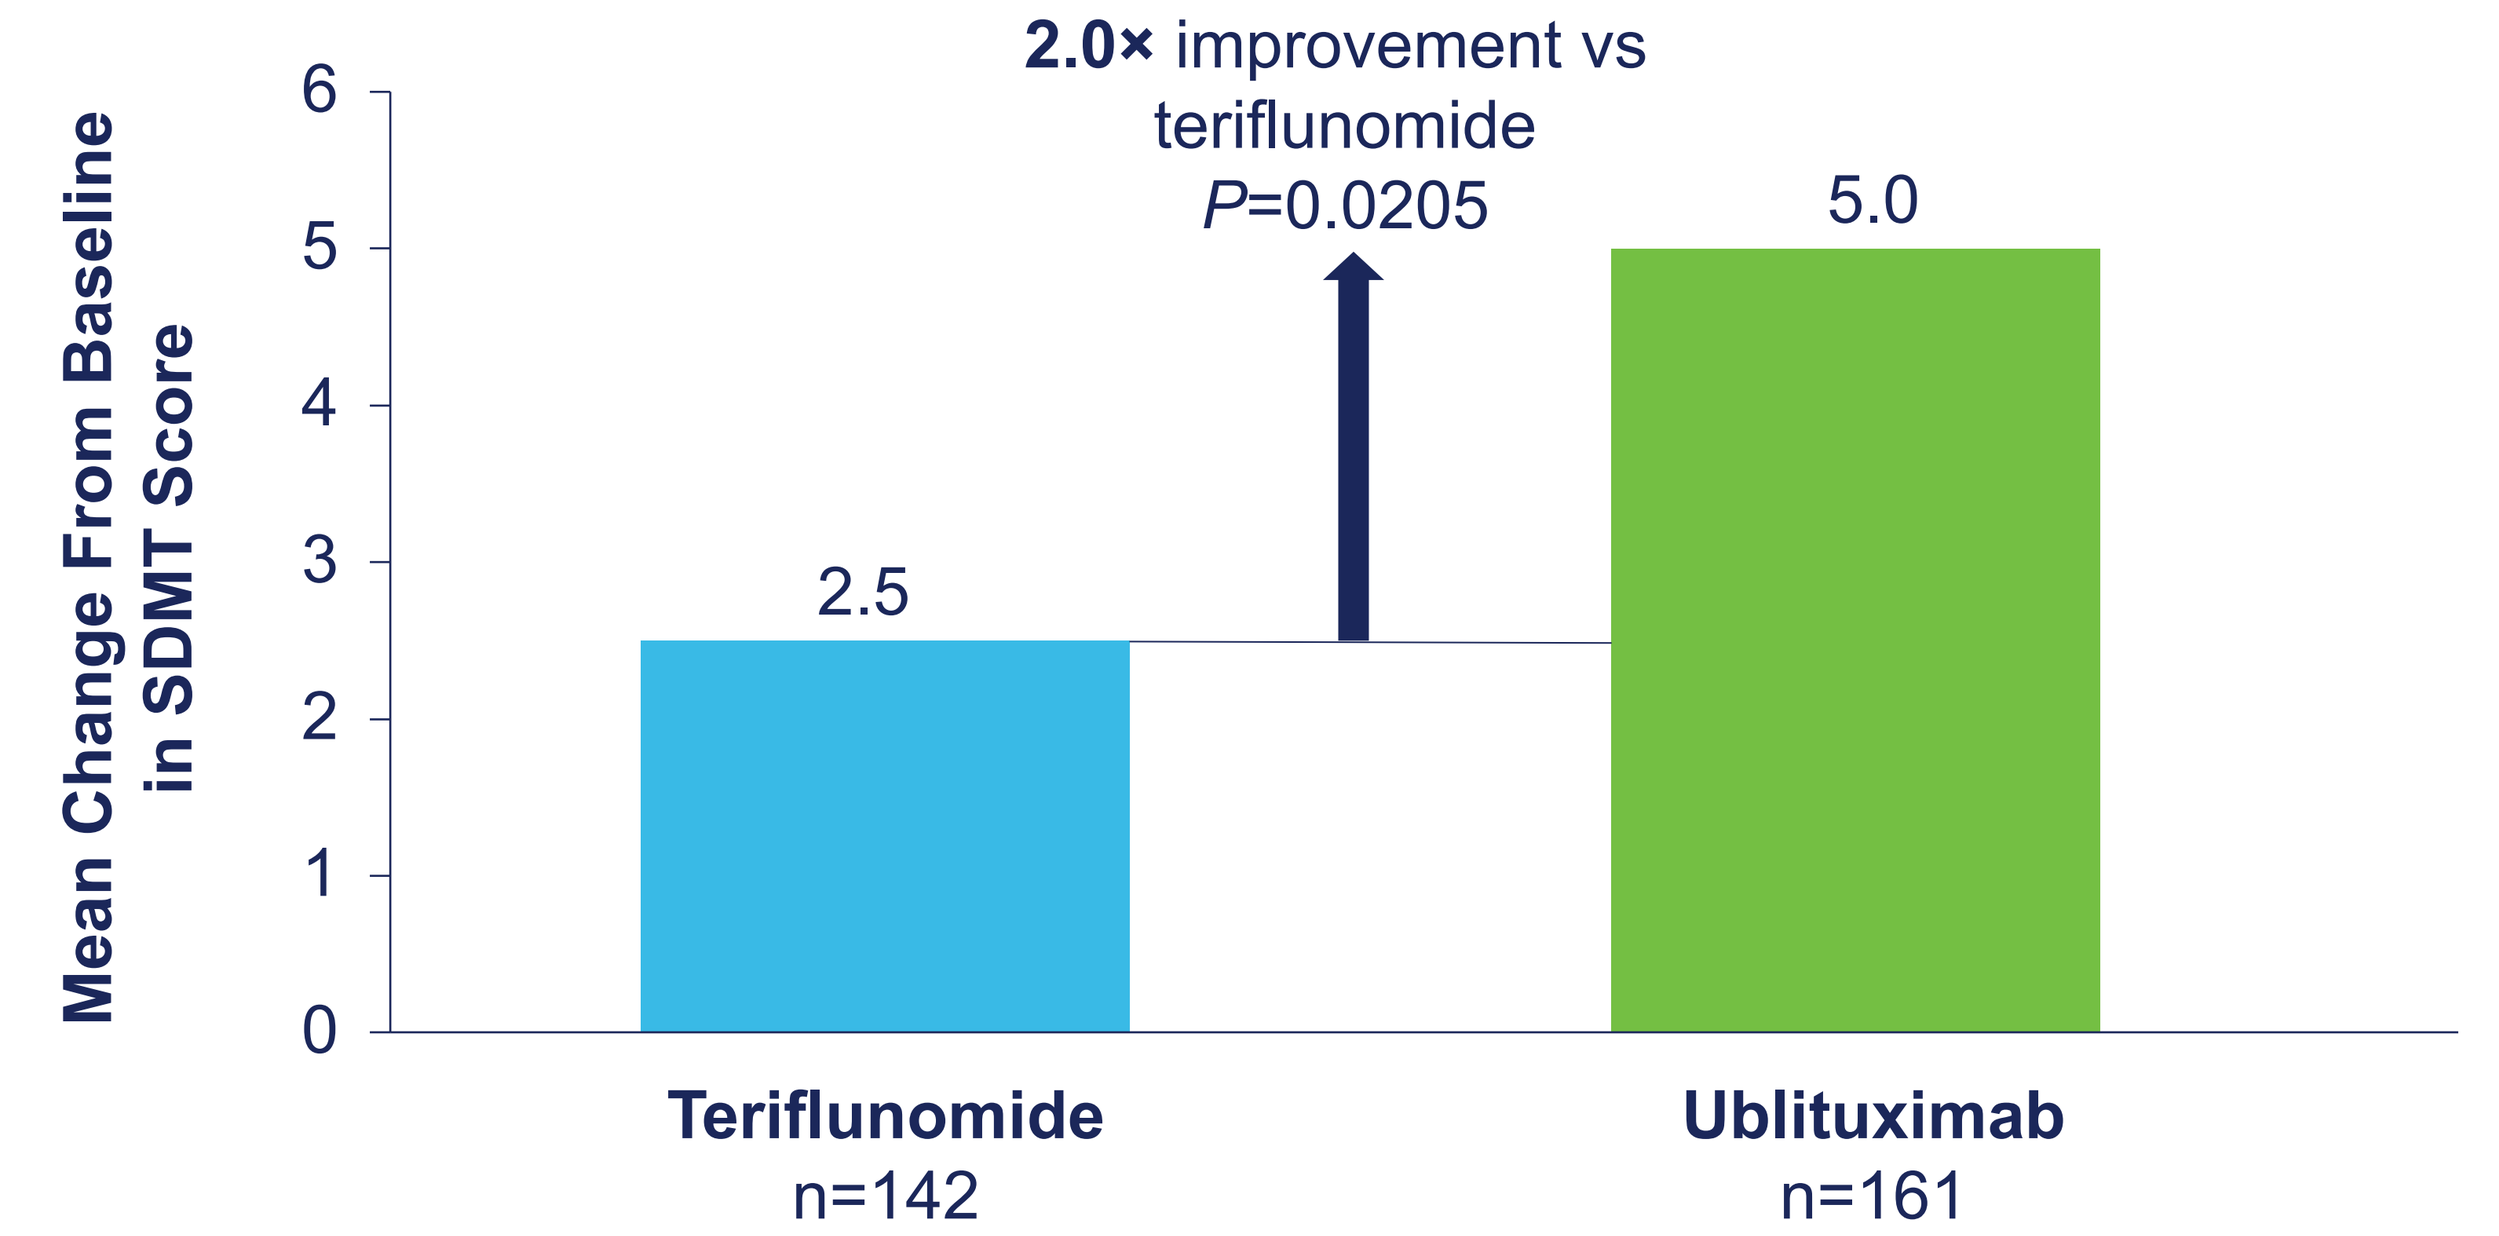
Mean Change From Baseline in SDMT Score
2.0× improvement vs teriflunomide
P=0.0205
2.5
5.0
Teriflunomide
n=142
Ublituximab
n=161

[a]Highly active disease at baseline defined as ≥1 relapse in prior year and either ≥1 Gd+ T1 lesion or ≥9 T2 lesions at baseline. Participants received approved disease-modifying therapy prior to enrollment.
Pooled post hoc analysis. mITT population. Gd+, gadolinium-enhancing; mITT, modified intention-to-treat; SDMT, Symbol Digit Modalities Test.

Presented at the 2022 Annual Meeting for The Consortium of Multiple Sclerosis Centers (CMSC), June 1-4, 2022, National Harbor, Maryland

Poster DMT19